COCHLEAR ABLATION EFFECTS ON COCHLEAR NUCLEUS CHEMISTRY

K. T. Robinson, D.A. Godfrey
K. Chen, M.A. Godfrey
Division of Otolaryngology & Dentistry,
Department of Surgery, University of Toledo
College of Medicine, Toledo, OH

ASTRACT and METHODS

Changes that occur in the posterior ventral cochlear nucleus (PVCN) following damage to the auditory nerve in the cochlea were measured. Chinchillas with cochlear ablations, as well as sham-lesioned chinchillas, were euthanized at times ranging from 3 to 84 days postablation. Both temporal bones and brains were saved. Temporal bones were fixed, embedded in paraffin and sectioned to document the completeness of the cochlear lesion. Brain portions containing the cochlear nuclei were frozen-sectioned, and sections were freeze dried. Freeze-dried sections were microdissected into samples of PVCN for high-performance liquid chromatography (HPLC) assay of glutamate and 11 other amino acid concentrations. The average glutamate concentration in the lesioned-side PVCN declined slightly by 7 days and by about 50% at 15 days; the decrease was maintained through 84 days post-ablation. Changes of glutamate concentration in the animals with the longer survival times correlated with the extent of cochlear damage. The density of PVCN tissue, measured as its dry weight per volume, showed no change through 31 days but declined by about 20% at 84 days post-ablation. The degeneration of the central portion of the auditory nerve following mechanical ablation of the cochlea is accompanied by decreases in glutamate concentration and density of PVCN tissue. These results are consistent with other evidence that damage to the cochlea leads to changes in the central auditory system. Although cochlear ablation is a more extreme lesion than would be encountered in the clinic, these results provide a baseline for studies that evaluate the effects of other types of cochlear damage on the central auditory system. Support by the American Tinnitus Association and the University of Toledo Foundation

INTRODUCTION

Information about sound is coded in the cochlea and carried to the central auditory system by the auditory nerve (CN VIII). The auditory nerve terminates entirely in the cochlear nucleus, the first brain center of the auditory system. The auditory nerve fiber terminals in the cochlear nucleus form a highly organized system of connections according to their peripheral innervation of the cochlea. The posterior ventral part of the cochlear nucleus (PVCN) contains a high density of auditory nerve fibers and terminals. Glutamate is the amino acid most likely to serve as the neurotransmitter of auditory nerve fibers in both the anterior (AVCN) and PVCN subdivisions of the ventral cochlear nucleus. Among cochlear nucleus regions, we focused our attention on the PVCN because, in our previous studies, it showed the largest decrease of glutamate concentration after cochlear ablation and the best correlation between glutamate and inner hair cell loss after ototoxic drug treatment.

RESULTS

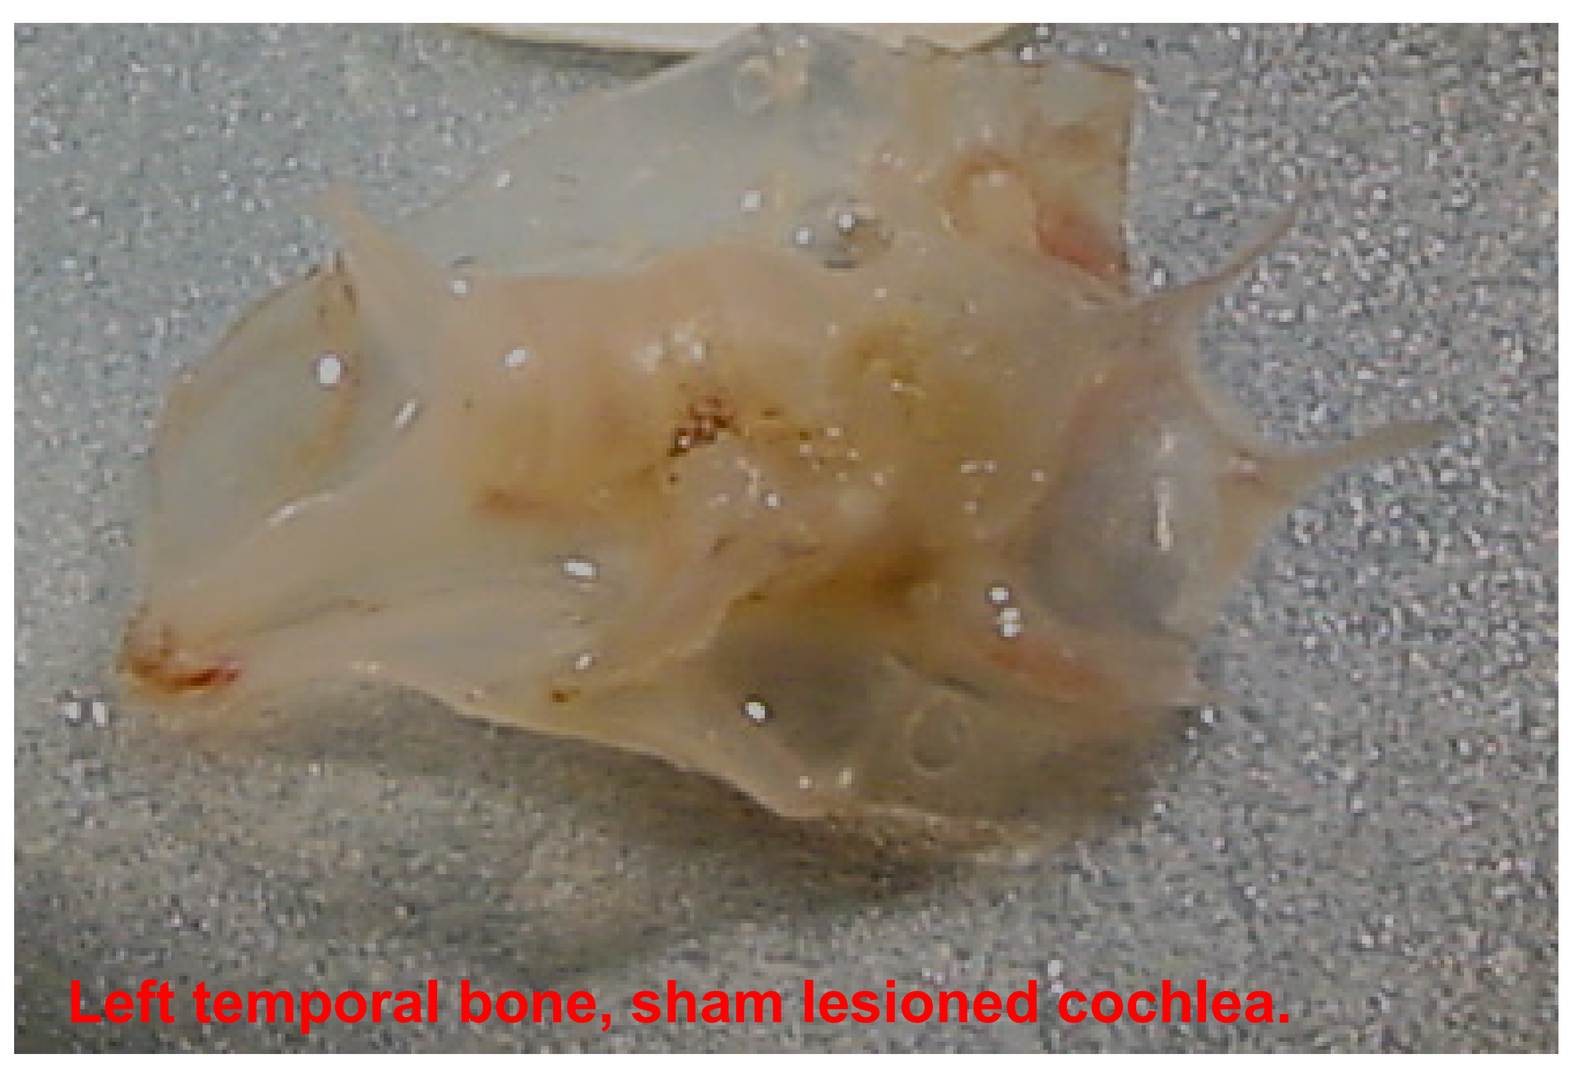

Left temporal bone, sham lesioned cochlea.

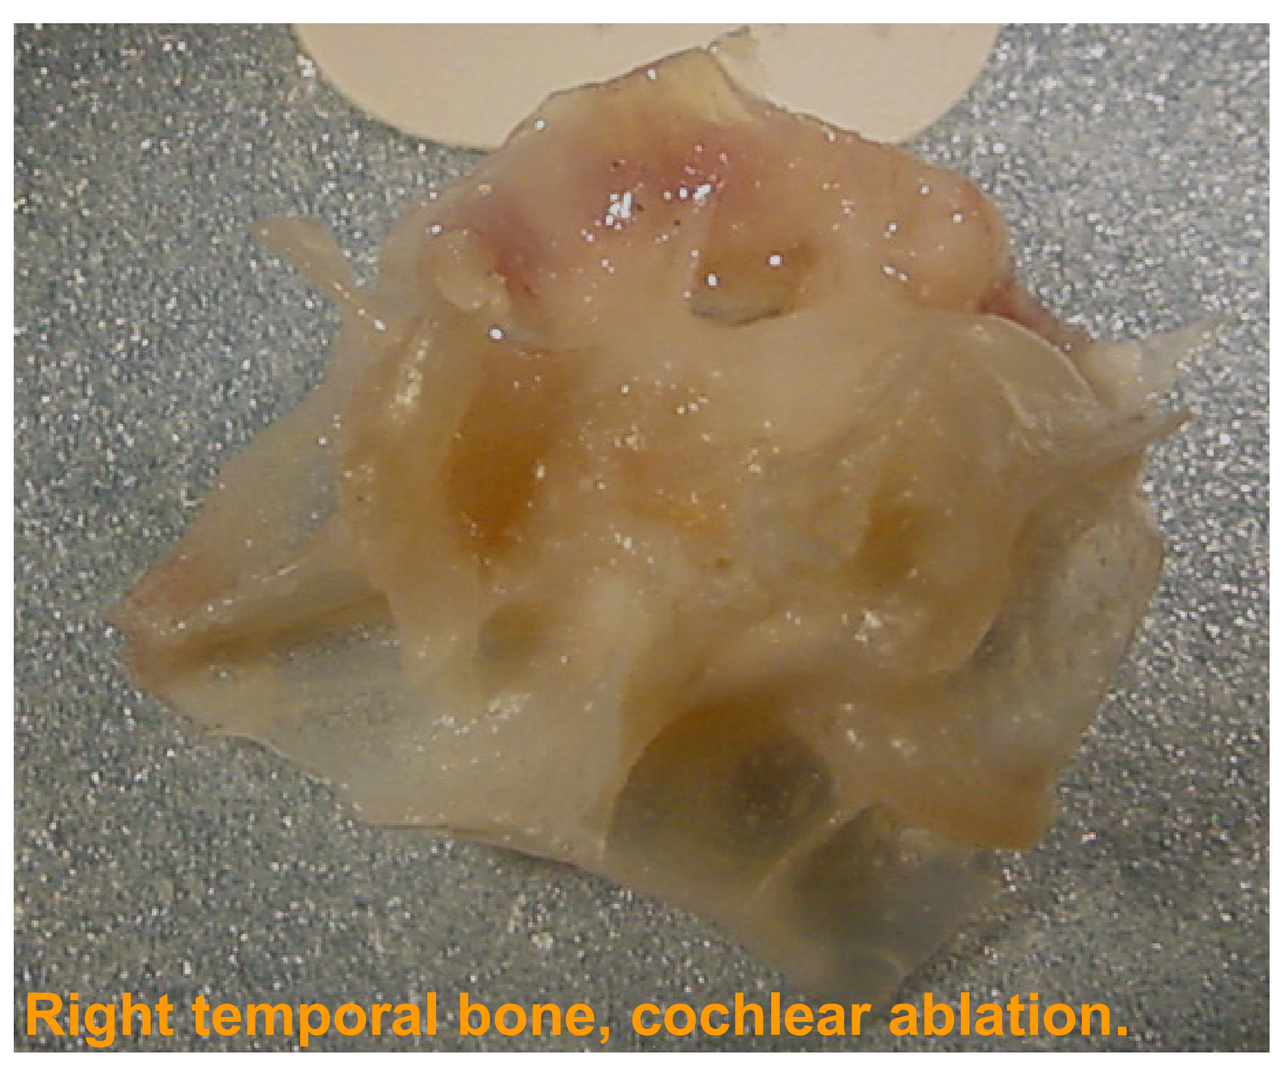

Right temporal bone, cochlear ablation.

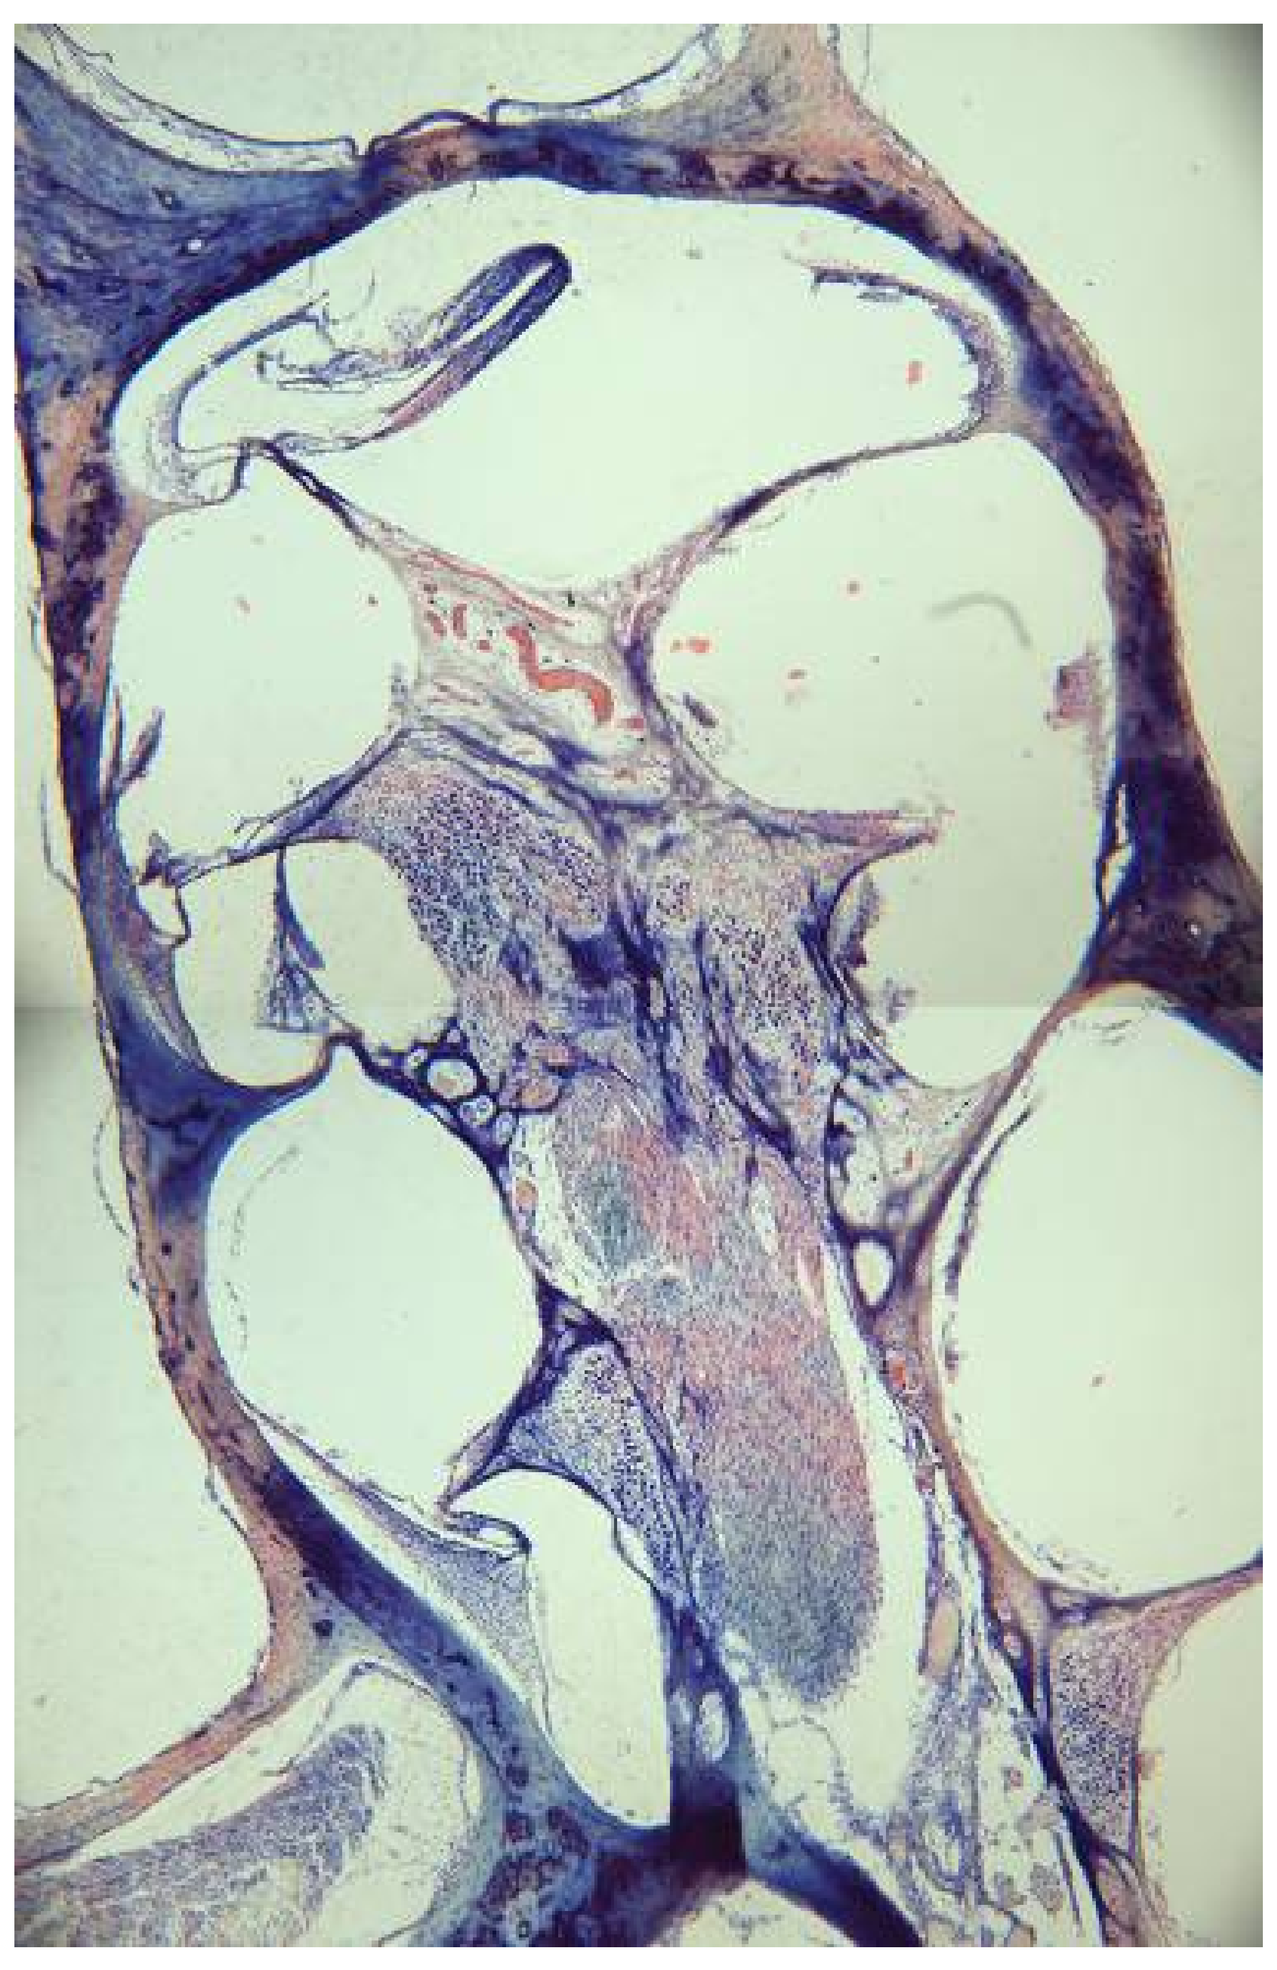

Section of left cochlea, sham lesioned. Normal spiral ganglion cells and no tissue debris within the scalae.

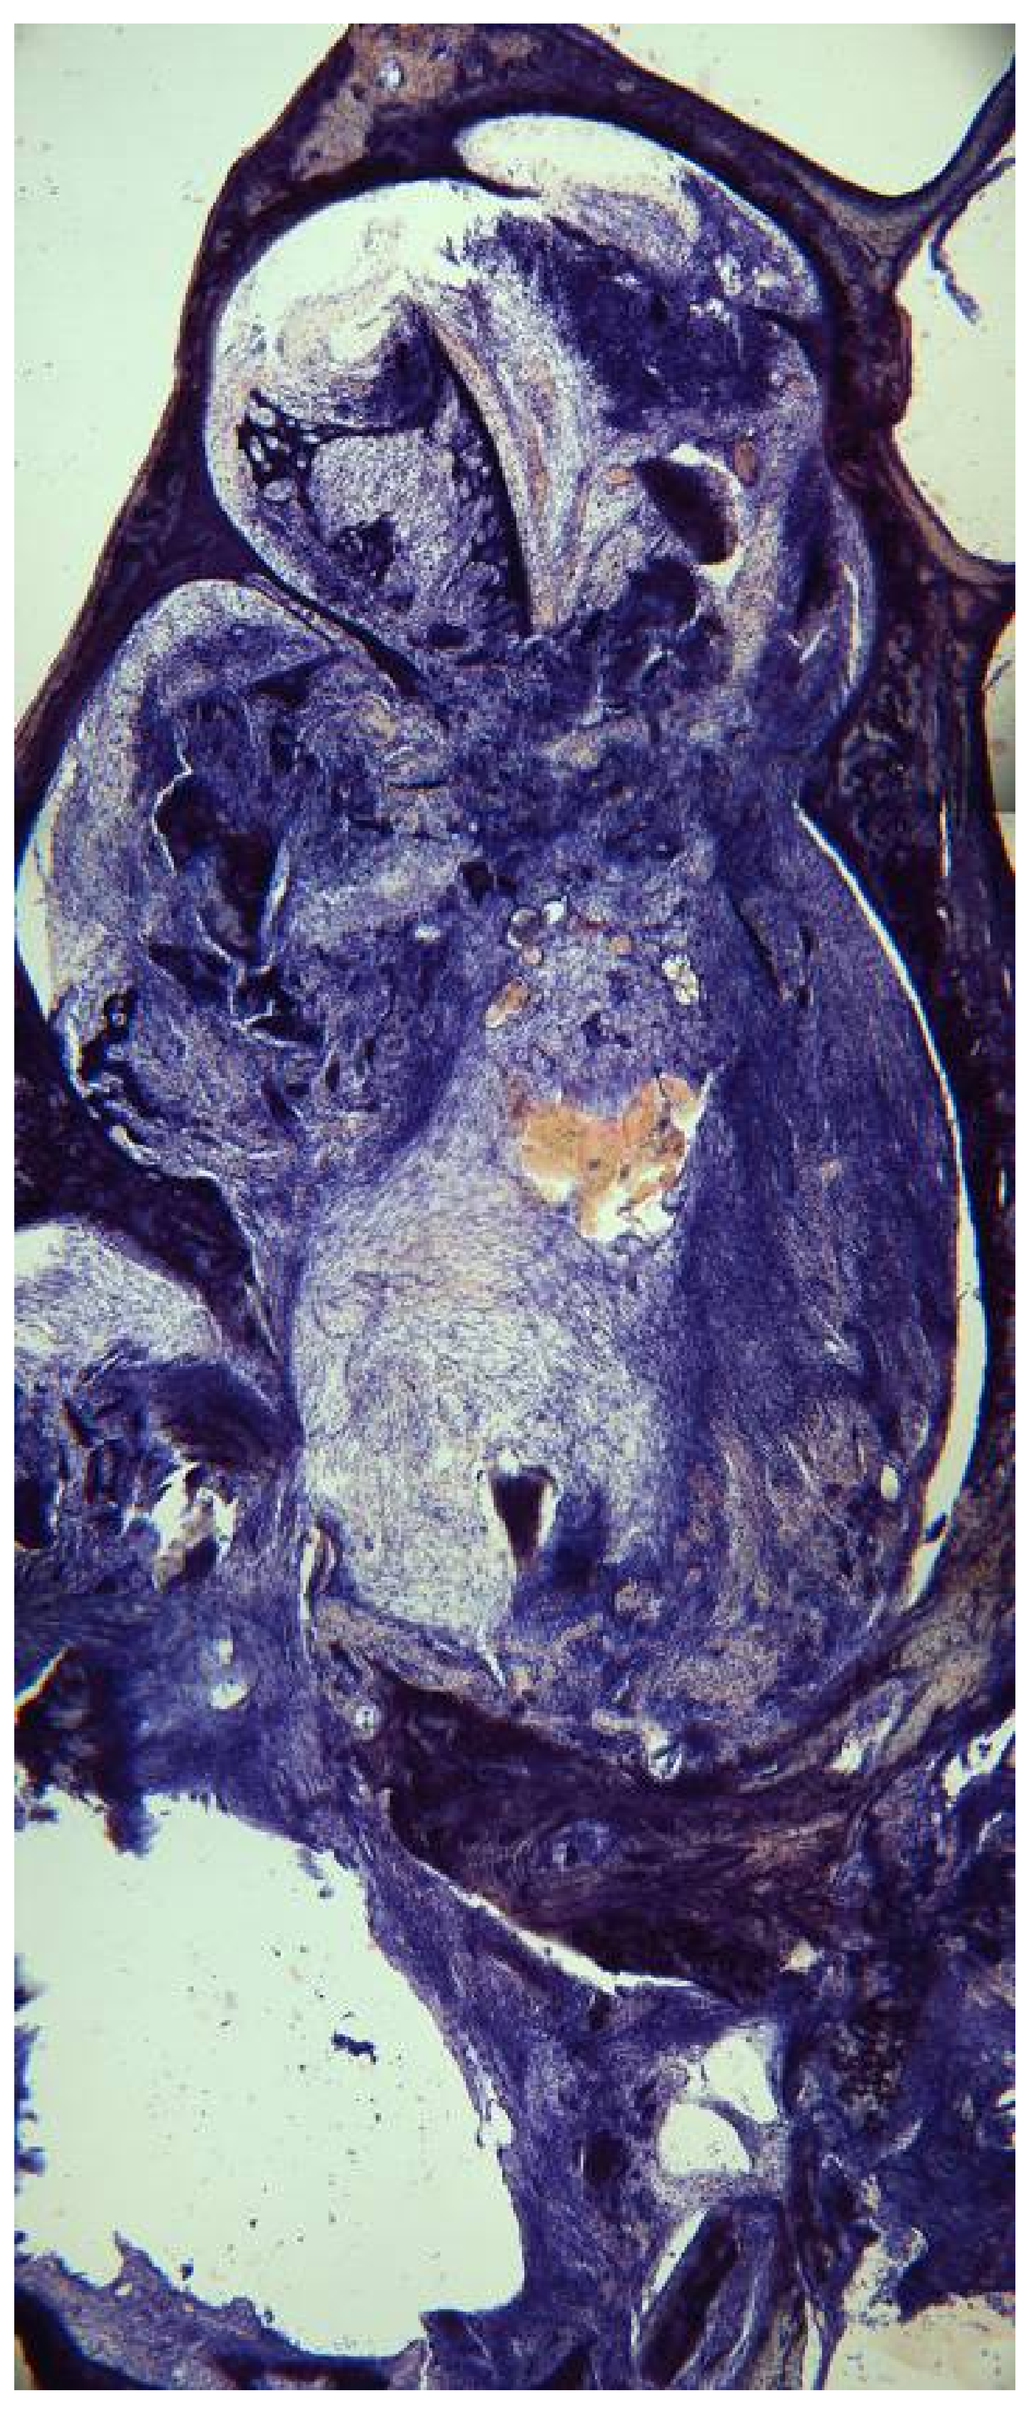

Section of right, ablated cochlea. Less spiral ganglion cells are visible and there is blood and tissue debris within the scalae.

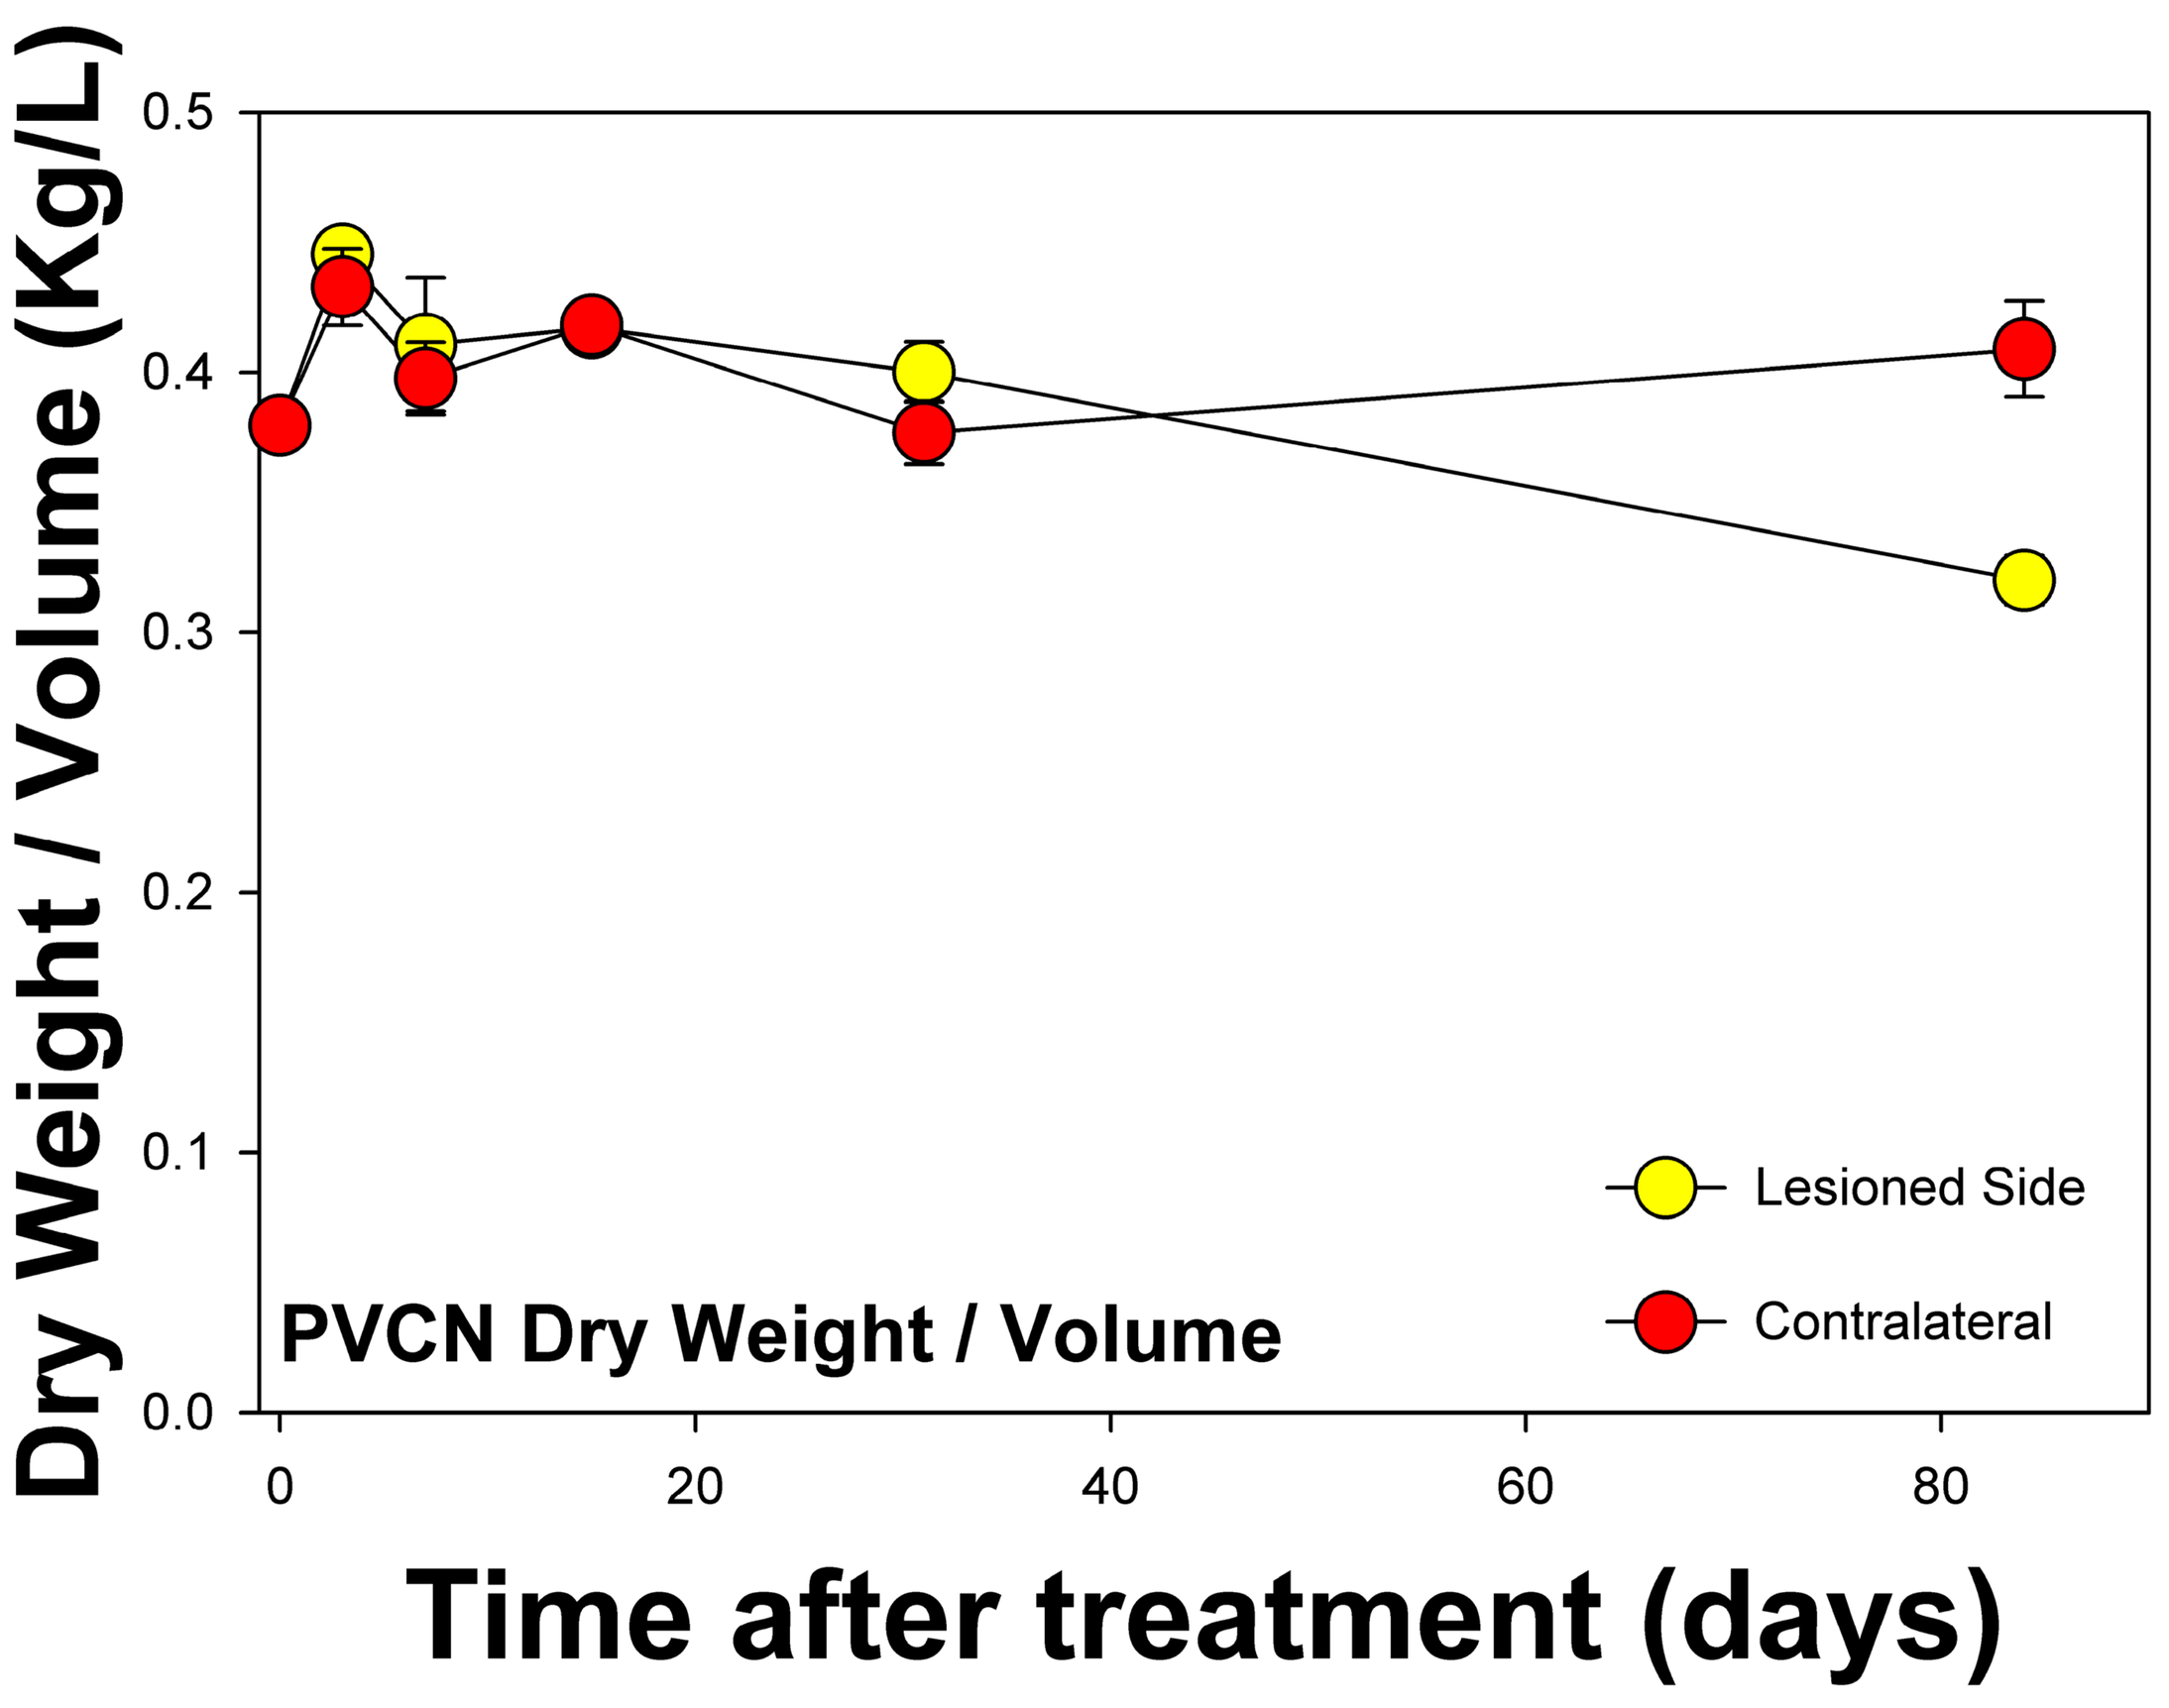
Dry Weight / Volume (Kg/L)
0.0
0.1
0.2
0.3
0.4
0.5
0
20
40
60
80
Lesioned Side
Contralateral
PVCN Dry Weight / Volume
Time after treatment (days)

The density of the PVCN tissue decreased by 84 days after cochlear ablation, but there was no clear change at any earlier time. Tissue density was measured as total dry weight of each PVCN section divided by its volume (area times section thickness). Each data point, with its standard error bar, is based on 4-8 measurements.

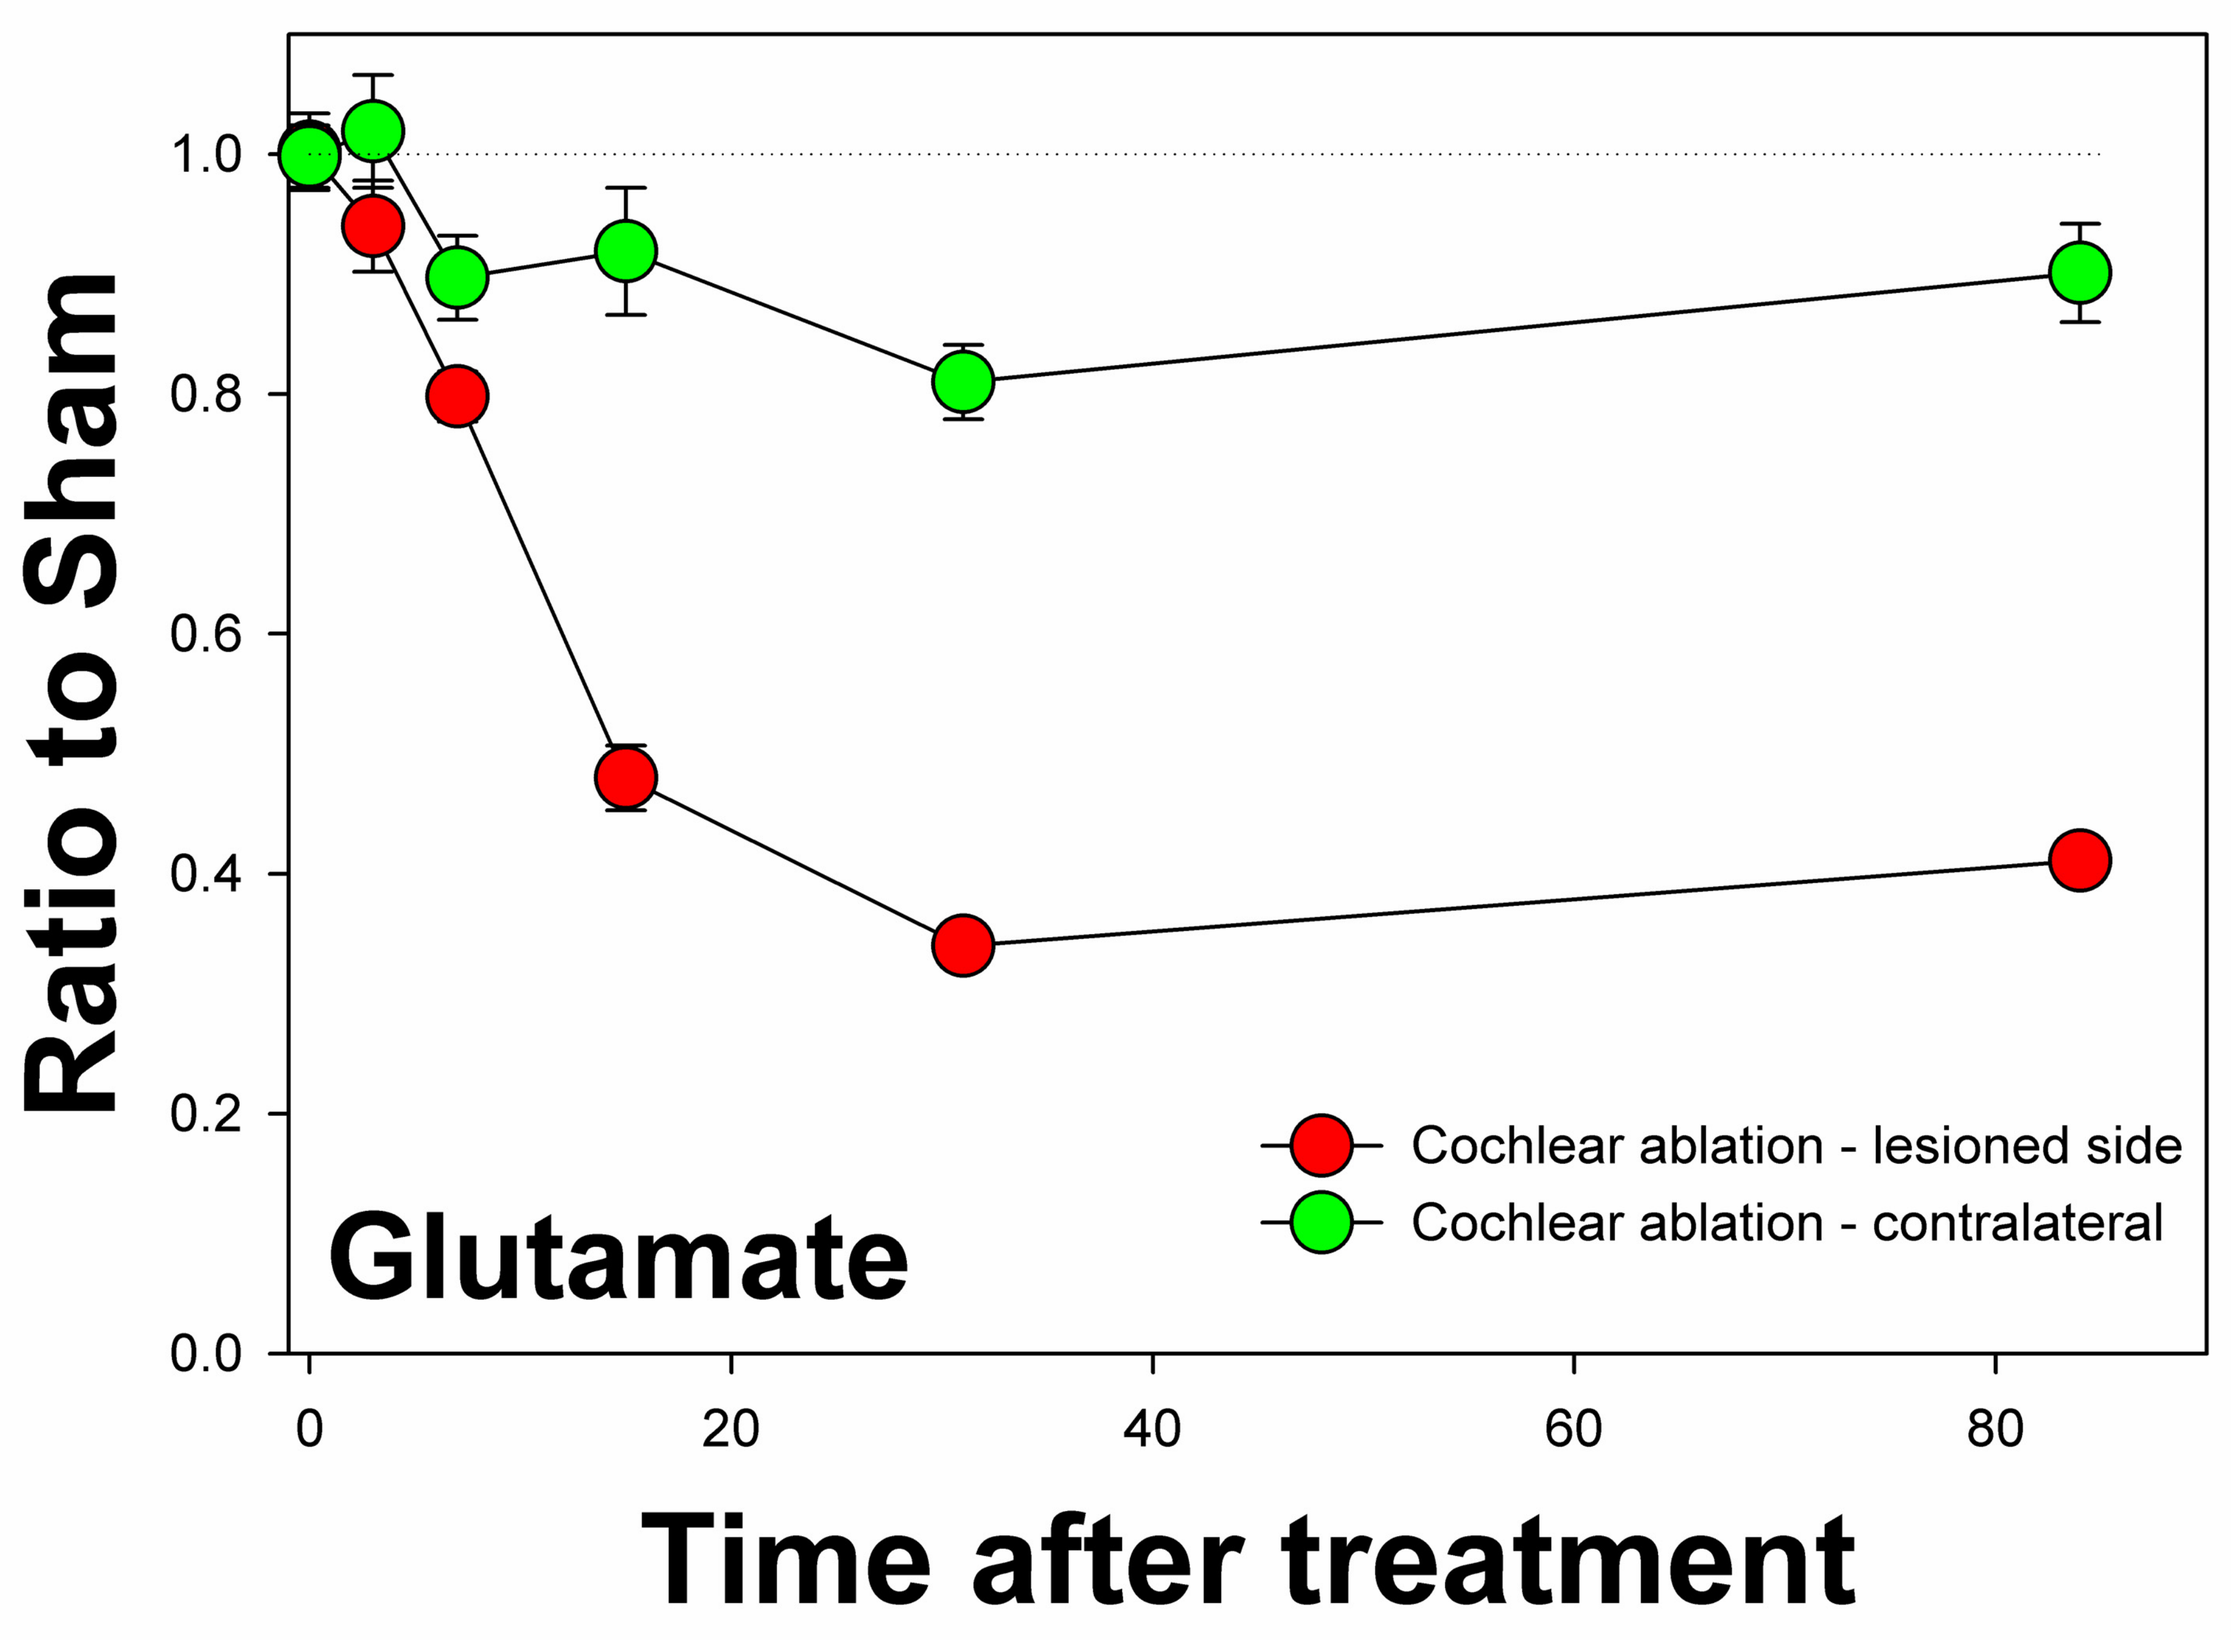
Ratio to Sham
0.0
0.2
0.4
0.6
0.8
1.0
0
20
40
60
80
Cochlear ablation - lesioned side
Cochlear ablation - contralateral
Glutamate
Time after treatment

The average glutamate concentration in the PVCN decreased significantly by 15 days after cochlear ablation. Each data point, with its standard error bar, is based on 7-30 samples. Dotted line represents ratio of 1.0.

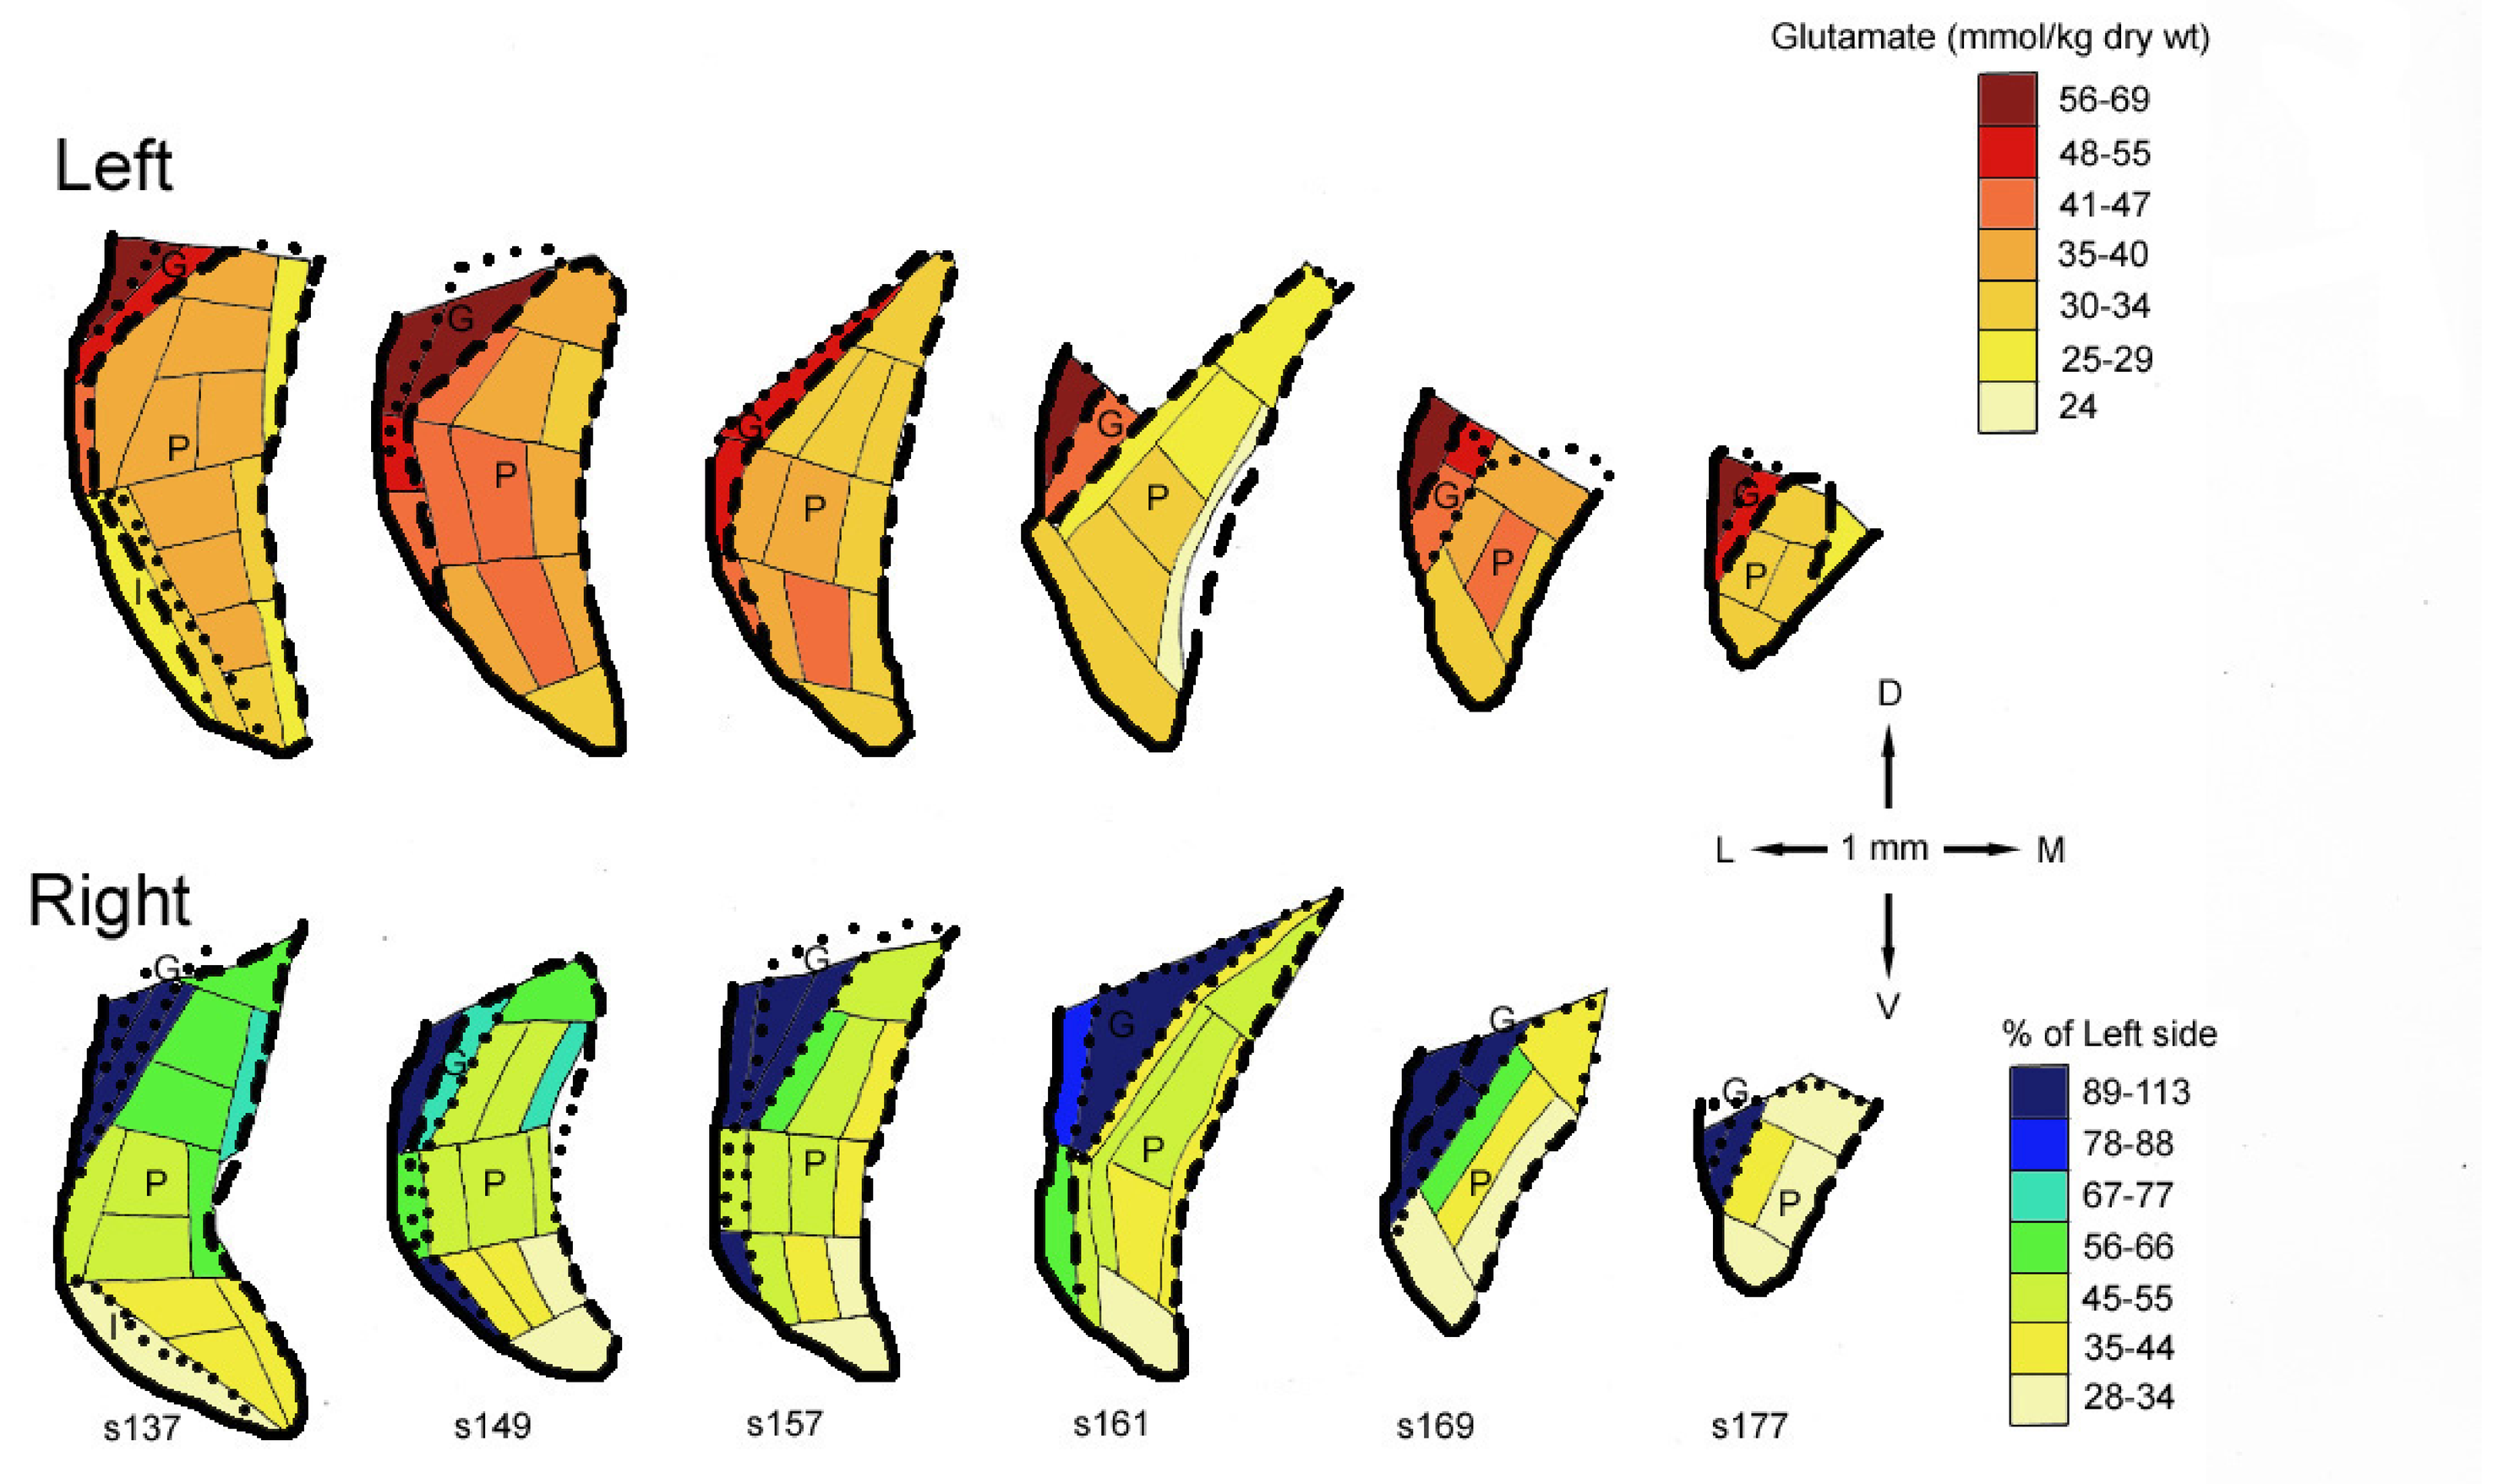
Glutamate (mmol/kg dry wt)
56-69
48-55
41-47
35-40
30-34
25-29
24
Left
Right
G
P
I
D
V
L
1 mm
M
% of Left side
89-113
78-88
67-77
56-66
45-55
35-44
28-34
s137
s149
s157
s161
s169
s177

The concentration of Glutamate in all parts of the right PVCN, except the granular regions, was greatly reduced at 31 days post ablation in a chinchilla. The glutamate concentrations for the samples are represented by color code for the **left** PVCN (**sham lesioned**). For the **right** PVCN (**ablated side**) and granular regions, glutamate concentrations are expressed as percentages of those for corresponding left-side samples, represented by a color code. Abbreviations are G, granular region; I, interstitial nucleus (auditory nerve root); P, PVCN. Directional scale shows dorsal (D), ventral (V), medial (M), and lateral (L) directions. Samples from left and right PVCN of the same section were dissected on the same day and assayed together.

CONCLUSIONS

- The gross appearance of the lesion to the cochlea was confirmed by microscopic examination of the sectioned cochlea. Loss of spiral ganglion cells was noted in lesioned cochleas.
- The tissue density of the lesioned-side PVCN showed no change up through 31 days but a significant decrease by 84 days post ablation.
- Glutamate concentration in the lesioned-side PVCN decreased significantly by 15 days post ablation and remained depressed through 84 days.
- Cochlear damage causes significant changes in cochlear nucleus neurotransmitter concentrations and PVCN tissue densities.